The Neural Correlates of Allocentric Spatial Memory Performance Across the Schizotypal Personality Spectrum

886.19

K. McMullen[1], E. Barkus[2], O. O'Daly[1], K. J. Craig[3], G. R. Dawson[3], C.T. Dourish[3], V. Kumari[4], S. C. Williams[1], & R. G. Morris[4]

(1) Centre for Neuroimaging Sciences, Institute of Psychiatry, King's College London, UK; (2) Department of Psychology, University of Wollongong, New South Wales, Australia; (3) **P1**vital, Department of Psychiatry, University of Oxford, Warneford Hospital, Oxford, UK;
(4) Department of Psychology, Institute of Psychiatry, King's College London, UK.

Introduction

- Schizotypy is a personality trait characterised by cognitive perceptual disturbances and impairments in social and cognitive functioning[1].
- Individuals with schizotypy demonstrate a range of cognitive deficits, as well as structural and functional differences across the brain, similar to those seen in schizophrenia but in an attenuated form.[2]
- Allocentric spatial memory relies on knowledge of the relationships between environmental cues, independent of one's own position[3]. It is dependent upon the integrity of the hippocampus and adjacent parahippocampal regions[4,5].
- Typically it has been measured in animals using standard preclinical tests such as the Morris Water Maze (MWM)[6] and in humans using virtual reality (VR) human analogues of this task.
- Behavioural[7] and functional MRI (fMRI)[8] studies have revealed impaired allocentric spatial memory performance in patients with schizophrenia.

Aims

- Using fMRI at 3T we sought to investigate the neural correlates of allocentric spatial memory in schizotypy using an VR human analogue of the MWM termed the Arena[5].

Methods

Participants

- 42 participants, aged 18-45, males and females, non or light (<5) smokers, fluent English.
- Average (M = 26.43±4.95) and High schizotypes (M = 48.14±4.90) as determined using the Schizotypal Personality Questionnaire (SPQ)[9]
- Exclusion criteria: Previous or current psychiatric disorder, substance use, neurological illness, visual or hearing impairment, migraines, MR incompatibility and consumption of prescribed or over the counter medication.

fMRI data acquisition:

- 3T GE HDx system. 38 axially oriented 3mm slices, TR/TE = 2s/3ms, flip angle 75, 64x64 matrix.
- Images were displayed on a Perspex screen at the bottom of the scanning bed.
- Navigation around the Arena was controlled using an MR compatible joystick.

References

1 Raine. Annu. Rev. Clin. Psychol. 2006. 2:291-326.
2 Siever et al. Schiz. Res. 2002. 54:151-167.
3 Burgess et al. Neuron. 2002. 35:625-641.
4 Morris et al. Nature. 1982. 297:681-683.
5 Parslow et al. Neuropsych. 2004. 18:450-461.
6 Morris. J. Neurosci. Methods. 1984. 11:47-60.
7 Hanlon et al. Schiz. Res. 2006. 87:67-80.
8 Folley et al. Neuroimage. 2009. 49: 3373-3384.
9 Raine. Schiz. Bull. 1991. 17:555-564.
10 Allen et al. Brain. Inj. 2007.21:1425-1428.
11 Callicott et al. Am. J. Psychiatry. 160:2209-2215.
12 Heckers et al. Nat Neuroscience. 1998. 1:318-323

Task Design

Layout:

Abstract patterns are rendered onto the walls of a circular VR arena, faded into one another to produce one large seamless pattern. Markers are randomly distributed on the floor of the arena to enhance perception of movement and perspective.

Procedure:

The task is divided into six epochs: Encoding, Rest (blank screen), Retrieval, Rest, Visual Control and Rest (Figure 1). Participants complete six trials during one fMRI session.

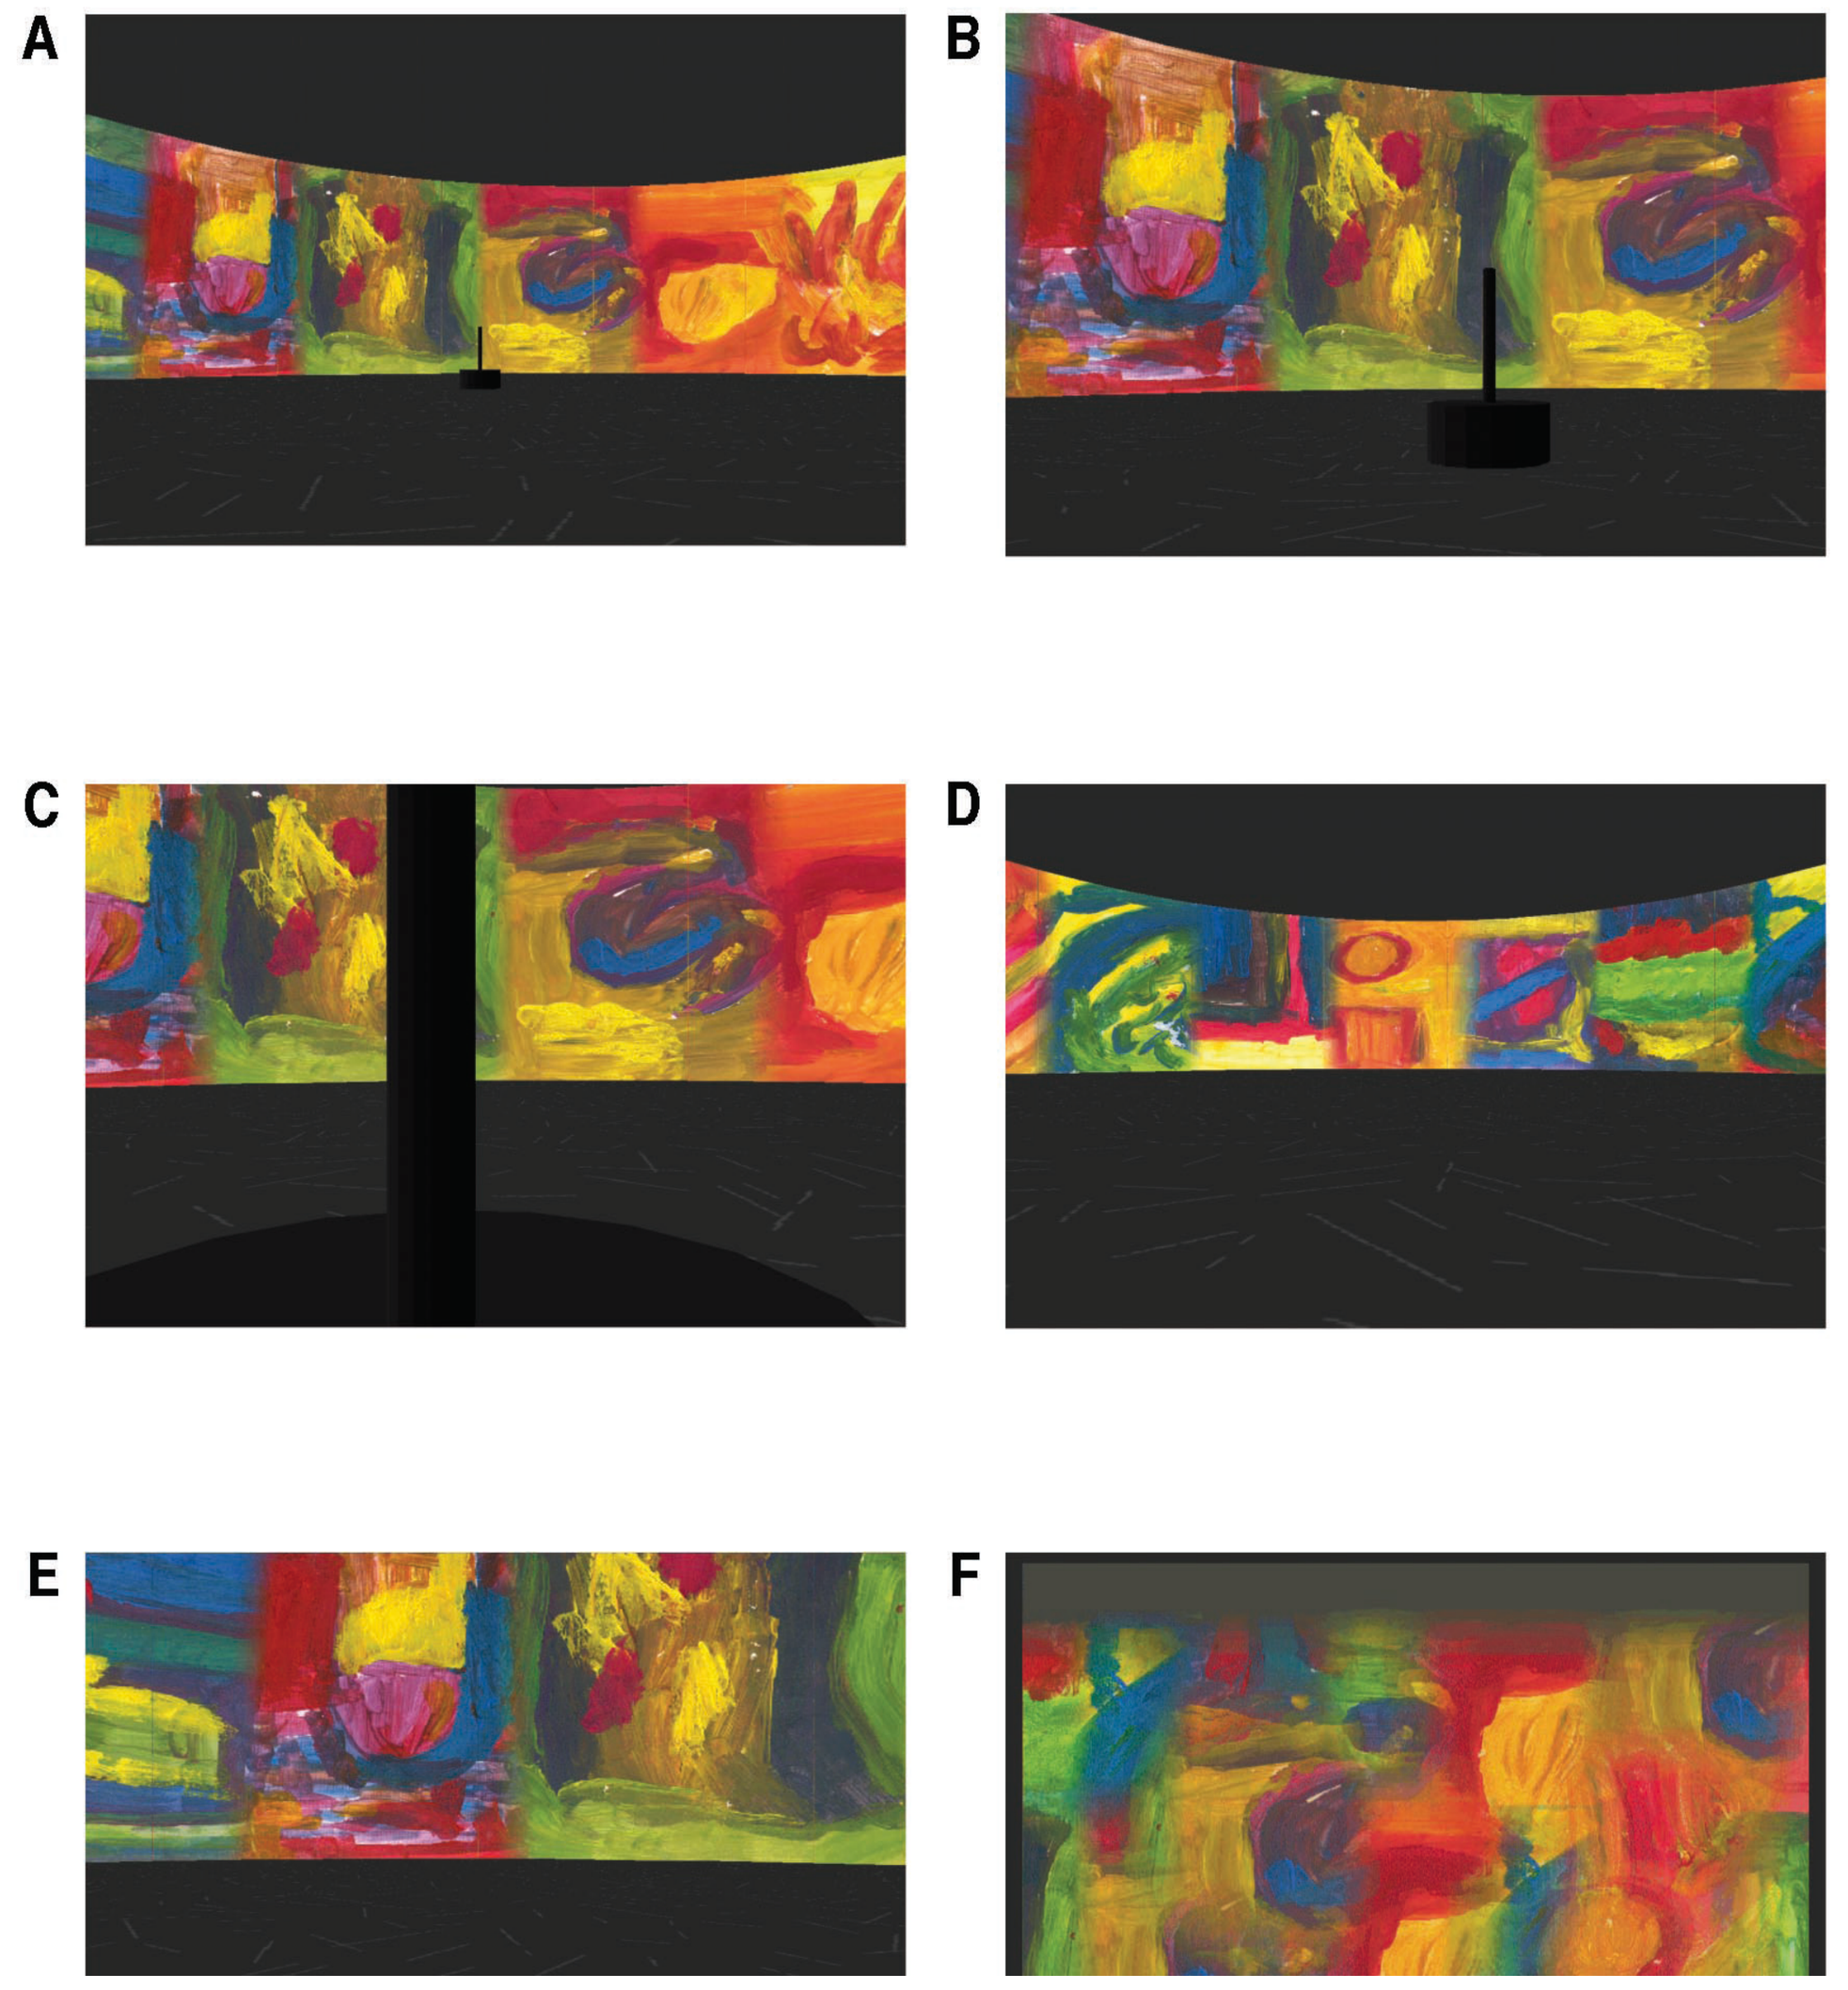
A
B
C
D
E
F

Figure 1.

Images from the Arena Task.

A. Encoding – participant sees pole in the distance.

B. Encoding – participant navigates towards the pole using the wall patterns to remember the pole location. C. Encoding – participant reaches the pole and stops.

D. Retrieval start position – different every time.

E. Retrieval finish position – the participant stops where they believe the pole was previously located.

F. Visual control condition.

Programmed in VR format by Third Dimension (Dorset, UK) using Superscape virtual reality software (Superscape, Hampshire, UK)

Task accuracy was defined as mean linear (distance between the true and estimated pole location) and angular deviation (difference between angle of true and estimated pole location).

Data Analysis:

- Behavioural data were analysed using independent t-tests in SPSS.
- FMRI data were pre-processed and analysed in SPM5 (statistical parametric mapping, UCL)
- Group differences were determined using a full factorial ANOVA, contrasting encoding and retrieval.
- *A priori* defined regions of interest were the hippocampus and parahippocampal formation.
- BOLD response was thresholded at $p < 0.001$ uncorrected.
- Significance defined as $p < 0.05$ FWE corrected for multiple comparisons.
- Small volume correction was applied using WFUpickatlas from the SPM5 toolbox.

Acknowledgements

Sincere thanks to David Gasston, the radiographers and the participants who took part in this study.
This study was supported by the **P1**vital CNS Experimental Medicine Consortium {members AstraZeneca, GlaxoSmithKline, Lundbeck, Organon (a subsidiary of Merck) and Pfizer}.

Reprint requests to: Katrina.McMullen@kcl.ac.uk

Results

Behavioural Results

Independent t-tests revealed no differences between groups in mean linear deviation ($t = 1.32$. $df = 40$, $p = 0.193$) or angular deviation ($t = -1.28$, $df = 40$, $p = 0.899$).

FMRI results

High schizotypes do not recruit the hippocampal-parahippocampal formation during encoding but demonstrate increased activation during memory retrieval relative to controls.

Figure 2: Hippocampal-Parahippocampal Formation (20, -12, 24, $p < 0.05$ FWE corrected)

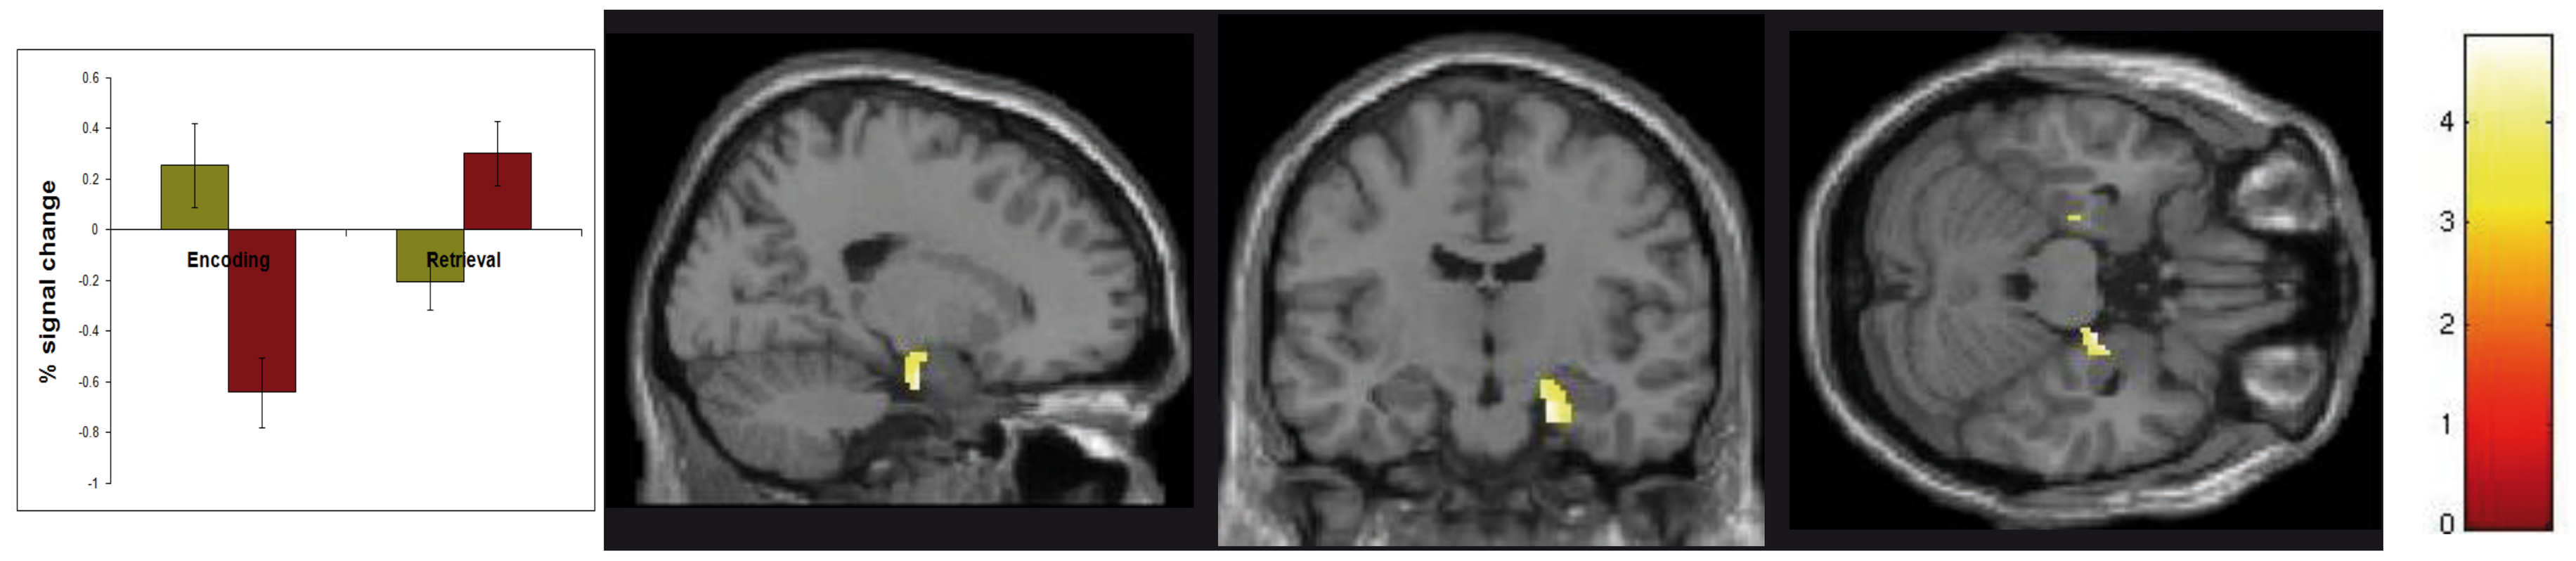
% signal change
Encoding
Retrieval
4
3
2
1
0

Figure 3: Hippocampal-Parahippocampal Formation (-20, 18, 20 $p < 0.05$ FWE corrected.

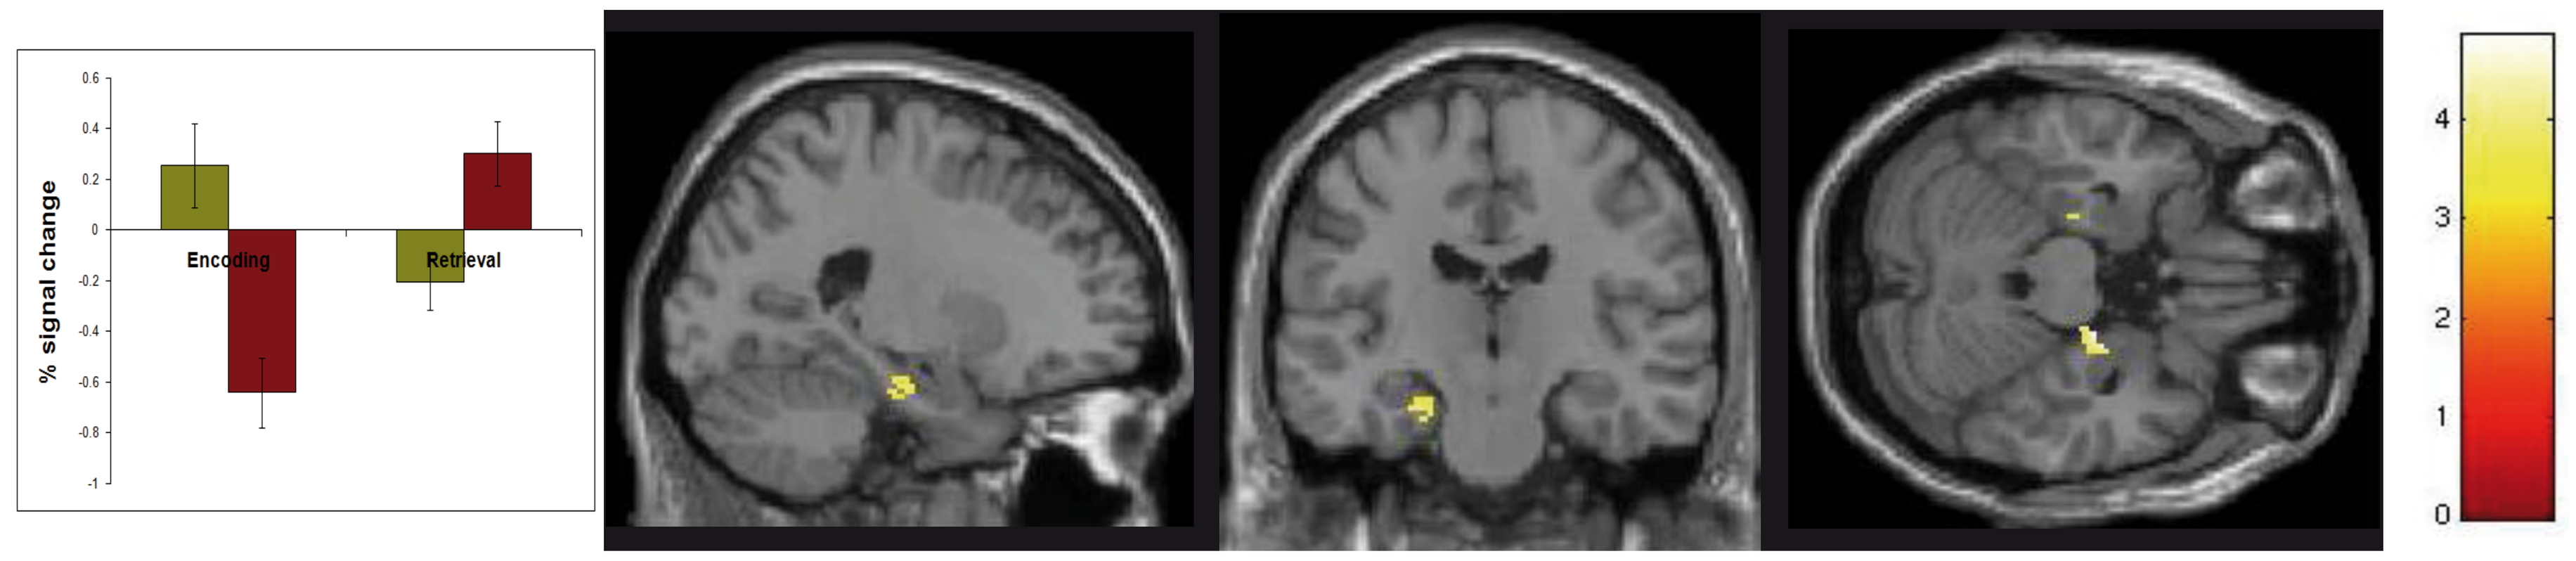
% signal change
Encoding
Retrieval
4
3
2
1
0

Additionally, high schizotypes had increased activation of the dorsolateral prefrontal cortex, inferior frontal gyrus and fusiform gyrus during memory retrieval.

Discussion

- High schizotypes employ a different neural strategy compared to average schizotypes and do not recruit the hippocampal-parahippocampal formation during encoding.
- Increased activation of the hippocampal parahippocampal formation during memory retrieval may indicate a sustained use of cognitive mapping as a compensatory mechanism.
- Increased activation of prefrontal regions may reflect increased cognitive effort as activation of the dorsolateral prefrontal cortex is associated with increased cognitive loadin memory tasks[10].
- These results are consistent with the schizophrenia literature[11,12] and support the idea of schizotypy as being on a continuum with schizophrenia.
- This study demonstrates the sensitivity of the arena task for detecting subtle neurocognitive differences in schizotypy.